CIVACIR, HEPATITIS C IMMUNE GLOBULIN (HCIG), POTENTLY NEUTRALIZES INFECTION OF HEPATITIS C VIRUS TRANSPLANT ESCAPE VARIANTS

Rajiv G. Tawar[1,2], Laura Heydmann[1, 2], Jörg Schüttrumpf[3], Shailesh Chavan[4], Mirjam B. Zeisel[1,2] and Thomas F. Baumert[1,2,5]

[1]Inserm, U1110, Institut de Recherche sur les Maladies Virales et Hépatiques, Strasbourg, France ; [2]University of Strasbourg, Strasbourg, France; [3]Biotest AG, Dreieich, Germany; [4]Clinical Research, Medical Affairs and Drug Safety, Biotest Pharmaceuticals, Boca Raton, Florida, United States;
[5]Pôle Hépato-digestif, Strasbourg University Hospital, Strasbourg, France

Introduction

- **Hepatitis C virus (HCV)-induced end-stage liver disease is the major indication of liver transplantation.**
- **Re-infection of liver graft is common.**
- **Safety and efficacy of new direct-acting antivirals (DAAs) for prevention of liver graft infection remains to be determined.**
- **Biotest Pharmaceuticals Civacir, a human hepatitis C antibody enriched immune globulin product (HCIG), has been shown to efficiently prevent liver graft infection in a phase III RCT (Terrault et al. EASL 2015).**

Aim of the study

- **We aimed to study the molecular mechanism of action of Civacir/HCIG against patient derived HCV escape variants.**

Methods

- **Inhibition of Civacir/HCIG-mediated HCV infection was studied using 22 viral variants isolated from patients before and after liver transplantation and state-of-the-art HCV cell culture models (Fafi-Kremer et al. J Exp Med 2010, Fofana et ål. Gastroenterology 2012; Felmlee, Fauvelle et al. EASL 2014).**
- **HCV pseudoparticles (HCVpp) and cell-culture-derived HCV (HCVcc) expressing patient-derived viral envelope glycoproteins from transplant escape variants were used.**

Civacir/HCIG potently and dose-dependently neutralizes escape and non-escape variants

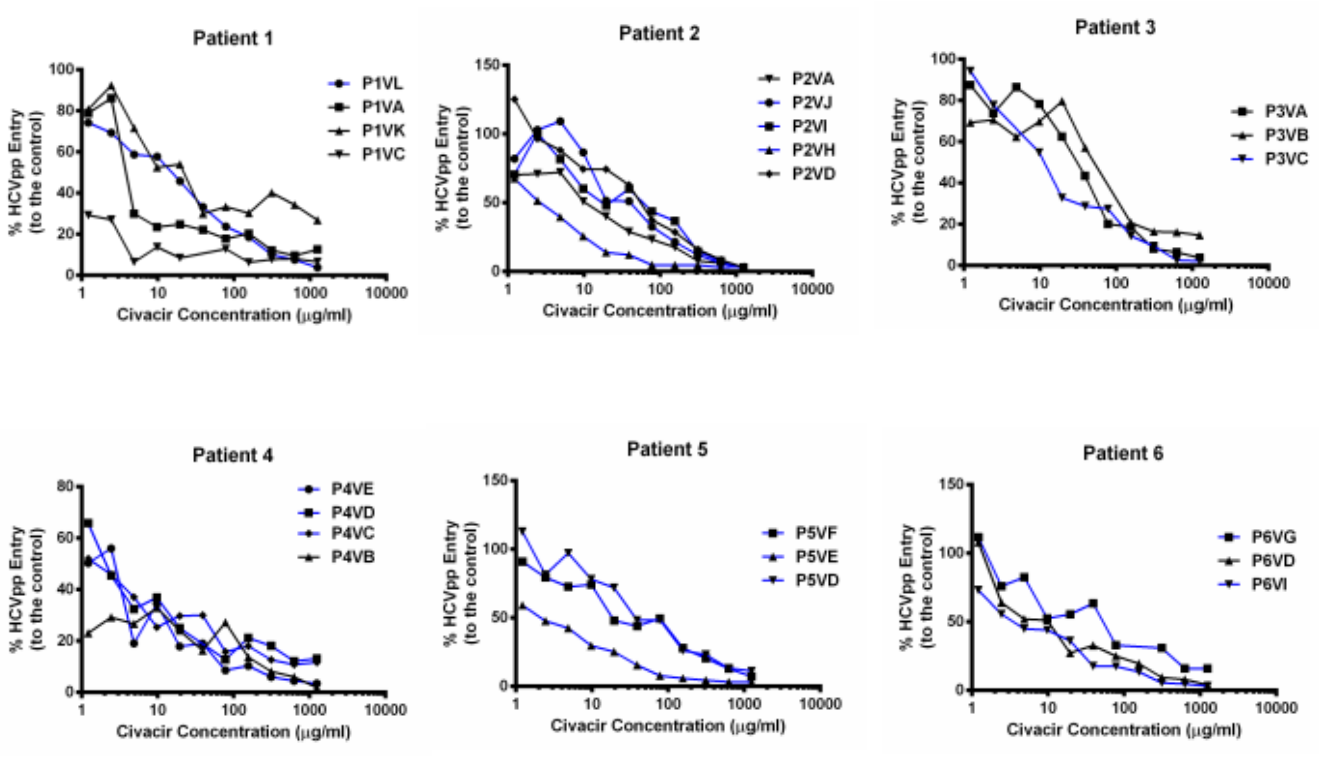
Patient 1
P1VL
P1VA
P1VK
P1VC
Patient 2
P2VA
P2VJ
P2VI
P2VH
P2VD
Patient 3
P3VA
P3VB
P3VC
Patient 4
P4VE
P4VD
P4VC
P4VB
Patient 5
P5VF
P5VE
P5VD
Patient 6
P6VG
P6VD
P6VI
% HCVpp Entry (to the control)
Civacir Concentration (µg/ml)

Figure 1. Civacir/HCIG potently and dose-dependently neutralizes HCVpp derived from escape and non-escape HCV variants isolated from patients before and after liver transplant. HCVpp expressing E1E2 envelope glycoproteins from escape (blue) and non-escape (black) variants isolated from 6 patients before and after liver transplantation respectively were produced and tested for neutralization against serially diluted Civacir/HCIG (1250 – 1.22 µg/ml). HCVpp were incubated with Civacir/HCIG or control IgG preparation at 37° C for 1 hour and subsequently inoculated on Huh7.5.1 hepatoma cells. The level of Infection was determined after 72 hours by measuring luciferase activity expressed as relative light unit (RLU). The level of HCVpp entry is shown as percentage of the control.

Civacir/HCIG equally neutralizes escape and non-escape variants at low nano-molar concentration

Patient no.	Viral variant	IC50 (µg/ml)	Correlation coefficient
Patient 1	P1VA	1.7	0.87
	P1VC	0.5	0.78
	P1VK	6.7	0.91
	P1VL	28.7	0.99
Patient 2	P2VA	22.0	0.98
	P2VD	36.6	0.95
	P2VH	3.3	1.00
	P2VI	85.8	0.88
	P2VJ	34.9	0.94
Patient 3	P3VA	37.9	0.97
	P3VB	96.1	0.92
	P3VC	10.4	0.97
Patient 4	P4VB	53.4	0.94
	P4VC	5.7	0.72
	P4VD	1.6	0.93
	P4VE	5.6	0.84
Patient 5	P5VD	34.8	0.95
	P5VE	8.9	0.99
	P5VF	38.8	0.94
Patient 6	P6VD	2.1	0.92
	P6VG	8.6	0.78
	P6VI	12.4	0.96

Figure 2. The IC_{50} values (µg/ml) of Civacir/HCIG against 22 HCV pseudoparticles expressing E1E2 envelope glycoproteins of viral isolates from different patient derived escape (blue) and non-escape (black) variants. The IC_{50} values and correlation coefficients were calculated by a nonlinear three parameter least squares analysis using GraphPad prism software.

Civacir/HCIG neutralizes infection of HCVpp expressing envelopes of escape variants in primary human hepatocytes

A.

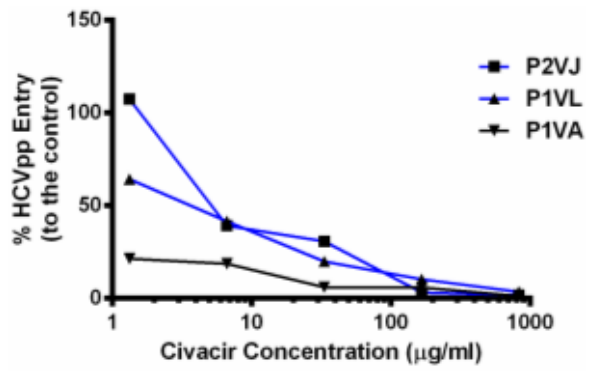
P2VJ
P1VL
P1VA
% HCVpp Entry (to the control)
Civacir Concentration (µg/ml)

B.

Viral variant	IC50 (µg/ml)	Correlation coefficient
P1VA	13.6	0.95
P1VL	8.2	0.99
P2VJ	1.8	0.96

Figure 3. Civacir/HCIG neutralizes HCVpp entry of escape and non-escape variants in primary human hepatocytes. (A) HCVpp derived from representative escape (blue line) and non-escape (black line) variants from liver transplant patients were incubated with different concentrations of Civacir/HCIG or control IgG preparation at 37° C for 1 hour and subsequently inoculated on primary human hepatocytes (PHH). The level of Infection was determined after 72 hours by measuring luciferase activity expressed as relative light unit (RLU). The level of HCVpp entry is shown as percentage of the control. (B) IC_{50} values of Civacir/HCIG against HCVpp derived from patient variants P2VJ, P1VL and P1VA. The IC_{50} values (µg/ml) were calculated by non-linear three parameter least squares analysis.

Civacir/HCIG neutralizes HCVcc expressing the envelope of a highly infectious escape variant (HCV P1VL)

A.

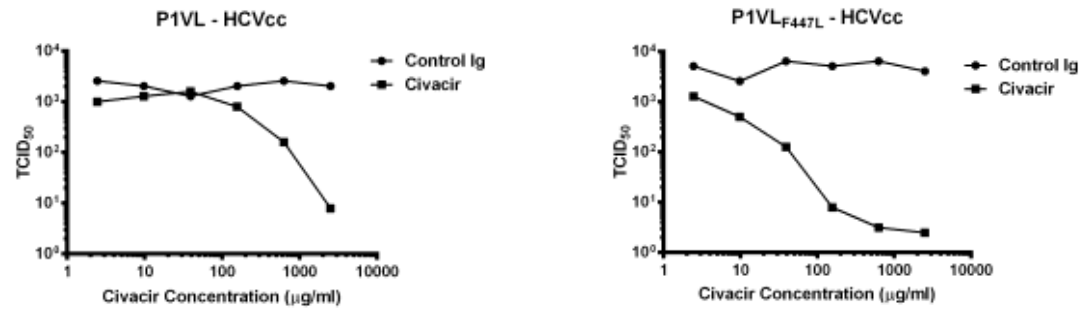
P1VL - HCVcc
P1VLF447L - HCVcc
Control Ig
Civacir
TCID50
Civacir Concentration (µg/ml)

B.

Viral variant	IC50 (µg/ml)	Correlation coefficient
$P1VL_{F447L}$	10.5	0.95
P1VL	375.0	0.85

Figure 4. Civavir/HCIG robustly neutralizes HCVcc JFH-P1VL and JFH-$P1VL_{F447L}$ chimera. (A) JFH-based HCVcc chimeras expressing structural proteins of patient isolates P1VL (genotype 1b) and $P1VL_{F447L}$ were produced as described before (Fofana et al. Gastroenterology 2012). Replacing phenylalanine at position 447 with leucine endows phenotype of non-escape variant i.e. less infectious and increased susceptibility to neutralization with autologous serum; akin to isolate P1VA. HCVcc was incubated with Civacir/HCIG or control IgG preparation at 37° C for 1 hour and subsequently inoculated on Huh7.5.1 hepatoma cells. Infection was read after 72 hours by measuring luciferase activity expressed as relative light unit (RLU). HCVcc infectivity was measured by determining the tissue culture infectious dose 50% ($TCID_{50}$). (B) IC_{50} (µg/ml) values of Civacir/HCIG against HCVcc JFH-P1VL and JFH-$P1VL_{F447L}$ chimera. The IC_{50} values were calculated by non-linear three parameter least squares analysis. JFH1 stands for highly infectious HCV strain isolated from a Japanese patient with fulminant hepatitis.

Conclusions

A.

B.

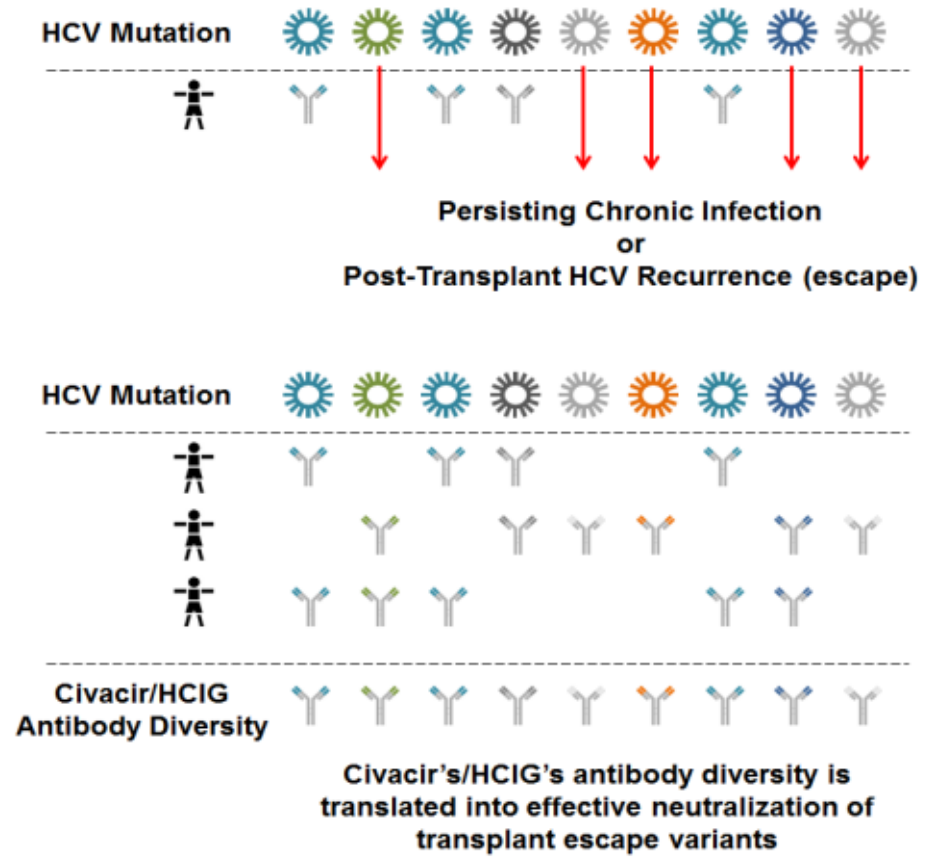
HCV Mutation
Persisting Chronic Infection
or
Post-Transplant HCV Recurrence (escape)
HCV Mutation
Civacir/HCIG
Antibody Diversity
Civacir's/HCIG's antibody diversity is
translated into effective neutralization of
transplant escape variants

Figure 5. HCV escape and antibody diversity. (A) The host antibody response is ineffective in countering the continuously evolving HCV quasispecies, leading to persistent chronic infection or escape and re-infection of liver graft post-transplantation. (B) Civacir, human hepatitis C antibody enriched immune globulin product (HCIG), is a polyclonal antibody preparation derived from human plasma enriched with HCV antibodies collected from several donors. The resulting high antibody diversity and synergy between anti-HCV antibodies targeting different epitopes is translated into effective neutralization of transplant escape variants resistant to autologous antibodies.

- **These results uncover the mechanism of action of Civacir/HCIG, explain its clinical efficacy for prevention of HCV liver graft infection and indicates its potential for use against different genotypes.**

This work was supported by the European Union (ERC-2008-AdG-233130-HEPCENT, INTERREG-IV-Rhin Supérieur-FEDER-Hepato-Regio-Net 2009 and 2012, HepaMab-FP7), ANRS, Laboratoire d'Excellence HepSYS (Investissement d'Avenir; ANR-10-LAB-28), Inserm and Université de Strasbourg.